EDP-305 Modulates Lipoprotein Metabolism Via Distinct Chromatin and miRNA Regulatory Mechanisms

Manuel Roqueta-Rivera, Mary Dinh Le Chau, Kelsey Garlick, Yang Li,
Guoqiang Wang, Yat Sun Or, and Li-Juan Jiang
Enanta Pharmaceuticals, Inc. Watertown, MA, USA

EASL The Home of Hepatology
THE INTERNATIONAL LIVER CONGRESS™ 2018
APRIL 11-15. PARIS. FRANCE

ENANTA Pharmaceuticals

Introduction

The Farnesoid X Receptor (FXR) has emerged as an attractive target for the treatment of NASH. EDP-305, a selective and potent small molecule FXR agonist, is currently in clinical development for the treatment of NASH. We previously demonstrated EDP-305 treatments resulted in differential effects on the expression of genes involved in lipoprotein metabolism (LDLR, SRB1) when compared to Obeticholic Acid (OCA). These differences may result in a more positive lipoprotein profile *in vivo* for EDP-305. In this study, we sought to characterize transcriptional and post-transcriptional regulatory mechanisms by which EDP-305 regulates LDLR and SRB1 expression.

Mechanisms Underlying LDLR and SRB1 Gene Expression Differences Between EDP-305 and OCA

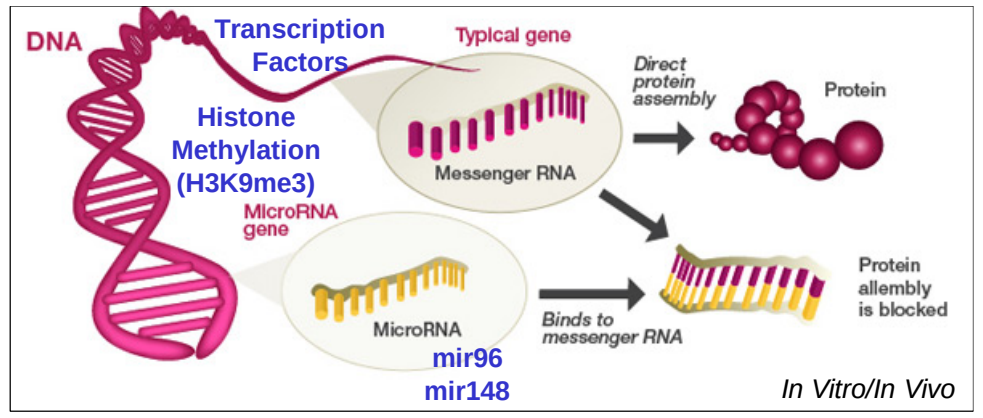
DNA
Transcription Factors
Typical gene
Histone Methylation (H3K9me3)
Messenger RNA
Direct protein assembly
Protein
MicroRNA gene
MicroRNA
mir96
mir148
Binds to messenger RNA
Protein allembly is blocked
In Vitro/In Vivo

Figure 1. Illustration depicting the transcriptional and post-transcriptional mechanisms by which EDP-305 may regulate LDLR and SRB1 gene expression. Methylation of Lysine-9 on Histone3 (H3K9me3) is a mark for gene repression. EDP-305 decreases H3K9me3 within the LDLR promoter and promotes increased recruitment of the transcription factor, sterol regulatory element binding protein 2 (SREBP2). At the SRB1 promoter, EDP305 increases H3K9me3. Additionally, EDP-305 regulates levels of microRNAs that target LDLR (miR 148) and SRB1 (miR 96).

Figure modified from http://www.longevinex.com/articles/micrornas-aging-and-resveratrol/

Materials and Methods

Cell Culture and Treatments. To examine the mechanisms of EDP-305 and OCA independent of drug potency, treatments were conducted near their respective EC50 concentrations (OCA @ 500nM; EDP-305 @ 50nM) in human hepatocytes (hepatoma and HepaRG).

Animal studies. C57B6/J mice were treated daily by oral gavage for 5 days with EDP-305 (30mpk) or OCA (100mpk).

MicroRNA (miRNA) isolation and Gene Expression. Total RNA and miRNAs were isolated from hepatocytes using the miRNAeasy kit (Qiagen). cDNA was prepared and gene expression was analyzed by real-time qPCR.

Protein. Liver tissue lysates (15-30 µg protein) were separated by 8% SDS-PAGE, transferred to nitrocellulose membrane, blocked with 5% nonfat dry milk and probed with antibodies against LDLR (Abcam) or SR-B1 (Novus Biologicals)**.**

Chromatin Immunoprecipitation (ChIP) -qPCR. Sonicated hepatocyte chromatin (~500bp) was used for ChIP assays to measure histone methylation and transcription factor enrichment at LDLR and SRB1 promoters.

EDP-305 Decreases H3K9me3 and Increases SREBP2 at the LDLR Promoter

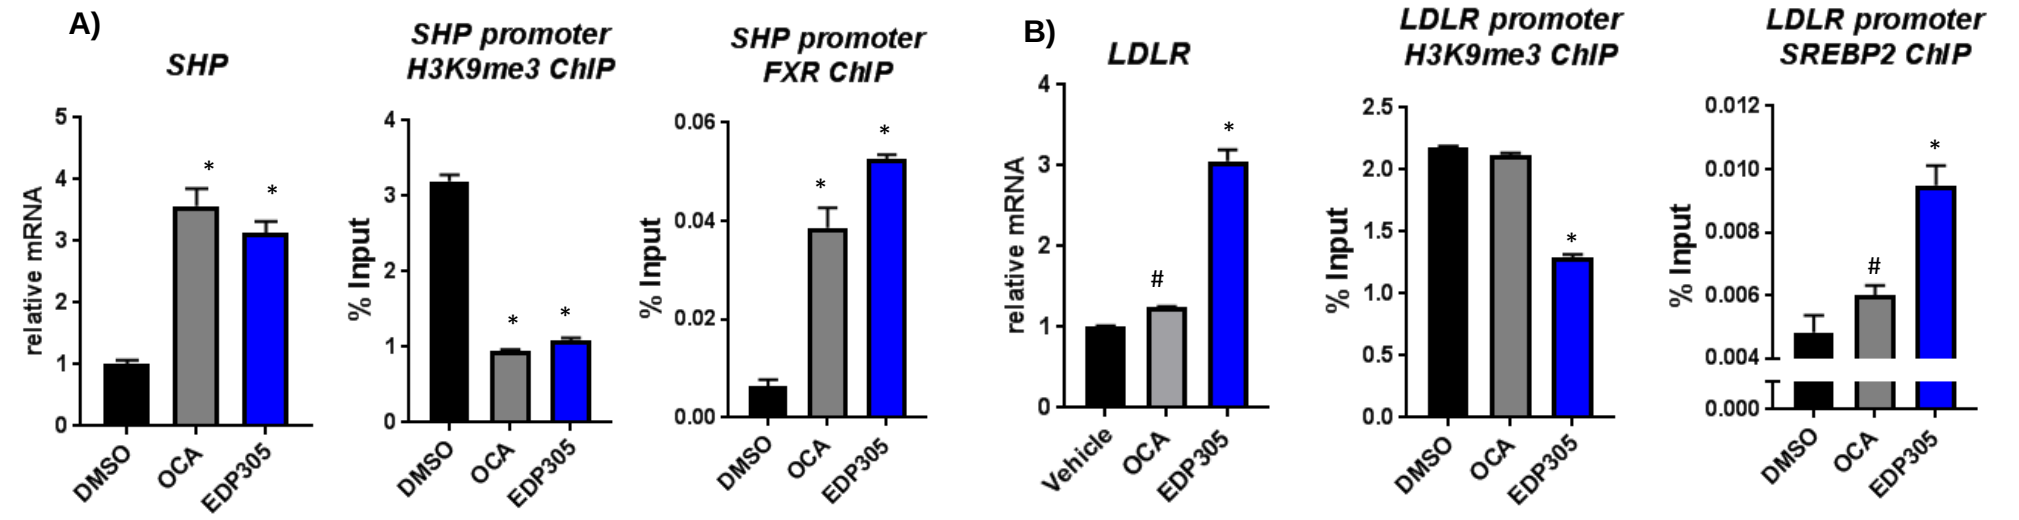
A)
SHP
SHP promoter H3K9me3 ChIP
SHP promoter FXR ChIP
B)
LDLR
LDLR promoter H3K9me3 ChIP
LDLR promoter SREBP2 ChIP
relative mRNA
% Input
DMSO
OCA
EDP305
Vehicle

Figure 2. Huh7.5 cells treated with EDP305 (50nM) or OCA (500nM) in presence of cholesterol. **A)** SHP mRNA and ChIP qPCR (H3K9me3, FXR) at SHP promoter. **B)** *LDLR* mRNA and ChIP qPCR (H3K9me3, SREBP2) at LDLR promoter. H3K9me3 is gene repressive mark. *p<0.05 relative to DMSO; #p<0.05 relative to EDP-305.

EDP-305 Increases H3K9me3 at the SRB1 Promoter

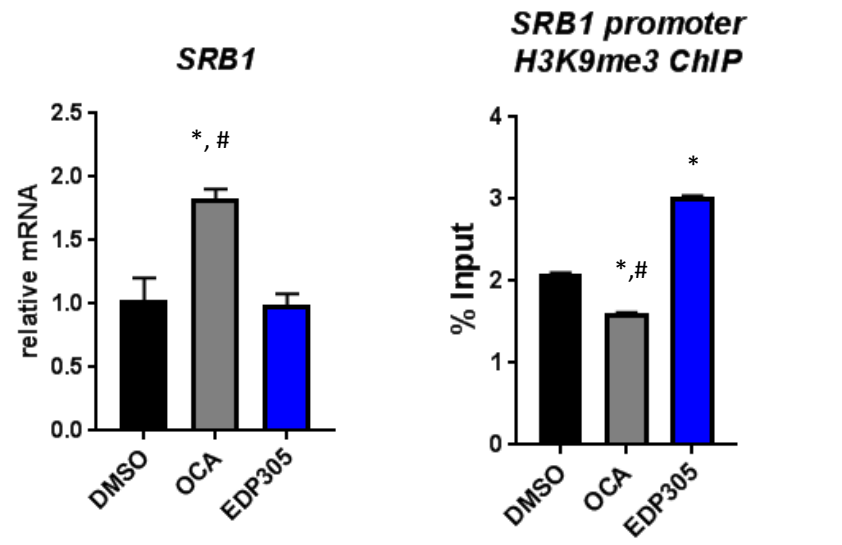
SRB1
SRB1 promoter H3K9me3 ChIP
relative mRNA
% Input
DMSO
OCA
EDP305

Figure 3. Huh7.5 cells treated with EDP305 (50nM) or OCA (500nM) in presence of cholesterol. *SRB1* mRNA and ChIP qPCR (H3K9me3) at SRB1 promoter. *p<0.05 relative to DMSO; #p<0.05 relative to EDP-305.

miRNAs that Target LDLR or SRB1

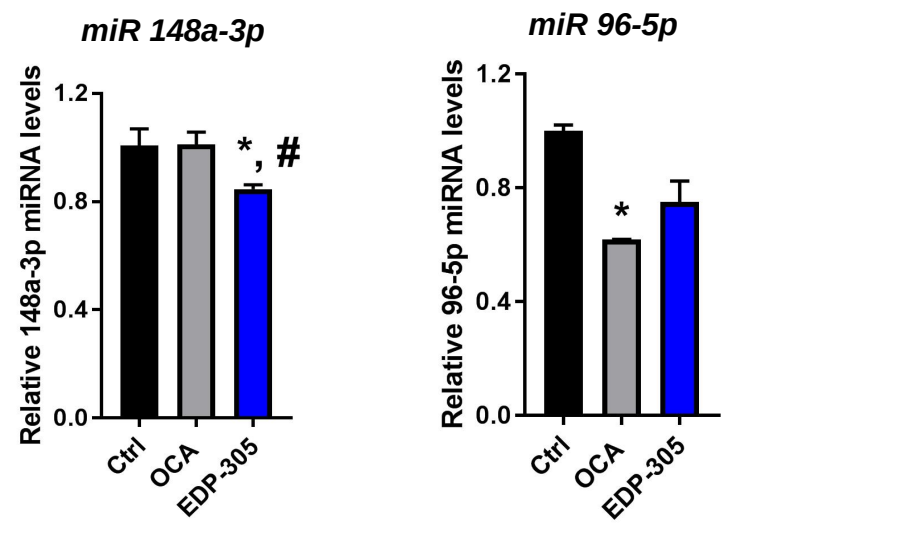
miR 148a-3p
miR 96-5p
Relative 148a-3p miRNA levels
Relative 96-5p miRNA levels
Ctrl
OCA
EDP-305

Figure 4. Relative miRNA levels in Huh7.5 cells treated with EDP305 (50nM) or OCA (500nM) in presence of cholesterol. miR148 degrades LDLR; miR96 degrades SRB1. *p<0.05 relative to DMSO; #p<0.05 relative to EDP-305.

miR 148a-3p and miR 96-5p Regulation by FXR Agonists in HepaRG

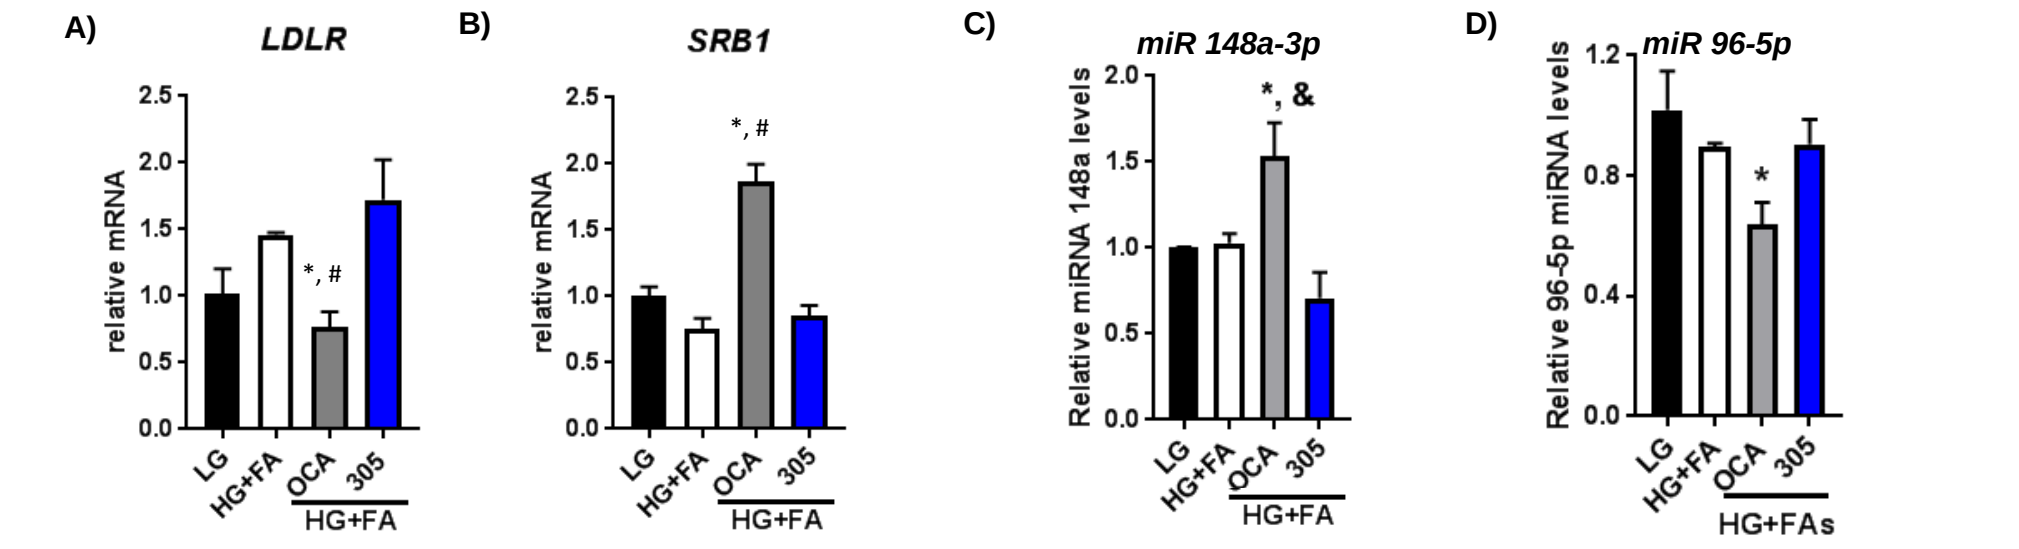
A)
LDLR
B)
SRB1
C)
miR 148a-3p
D)
miR 96-5p
relative mRNA
Relative miRNA 148a levels
Relative 96-5p miRNA levels
LG
HG+FA
OCA
305
HG+FA
HG+FAs

Figure 5. A and B) Relative *LDLR* and *SRB1* mRNA expression in HepaRG cells, treated with high glucose and fatty acids. **C and D**) Relative expression of miRs 148a-3p and miR 96-5p in HepaRG cells. *p<0.05 relative to DMSO; #p<0.05 relative to EDP-305. LG=Low Glucose; HG+FA=High Glucose with 200uM BAS conjugated palmitate and oleate.

EDP-305 Increases LDLR and Does Not Increase SRB1 mRNA *In Vivo*

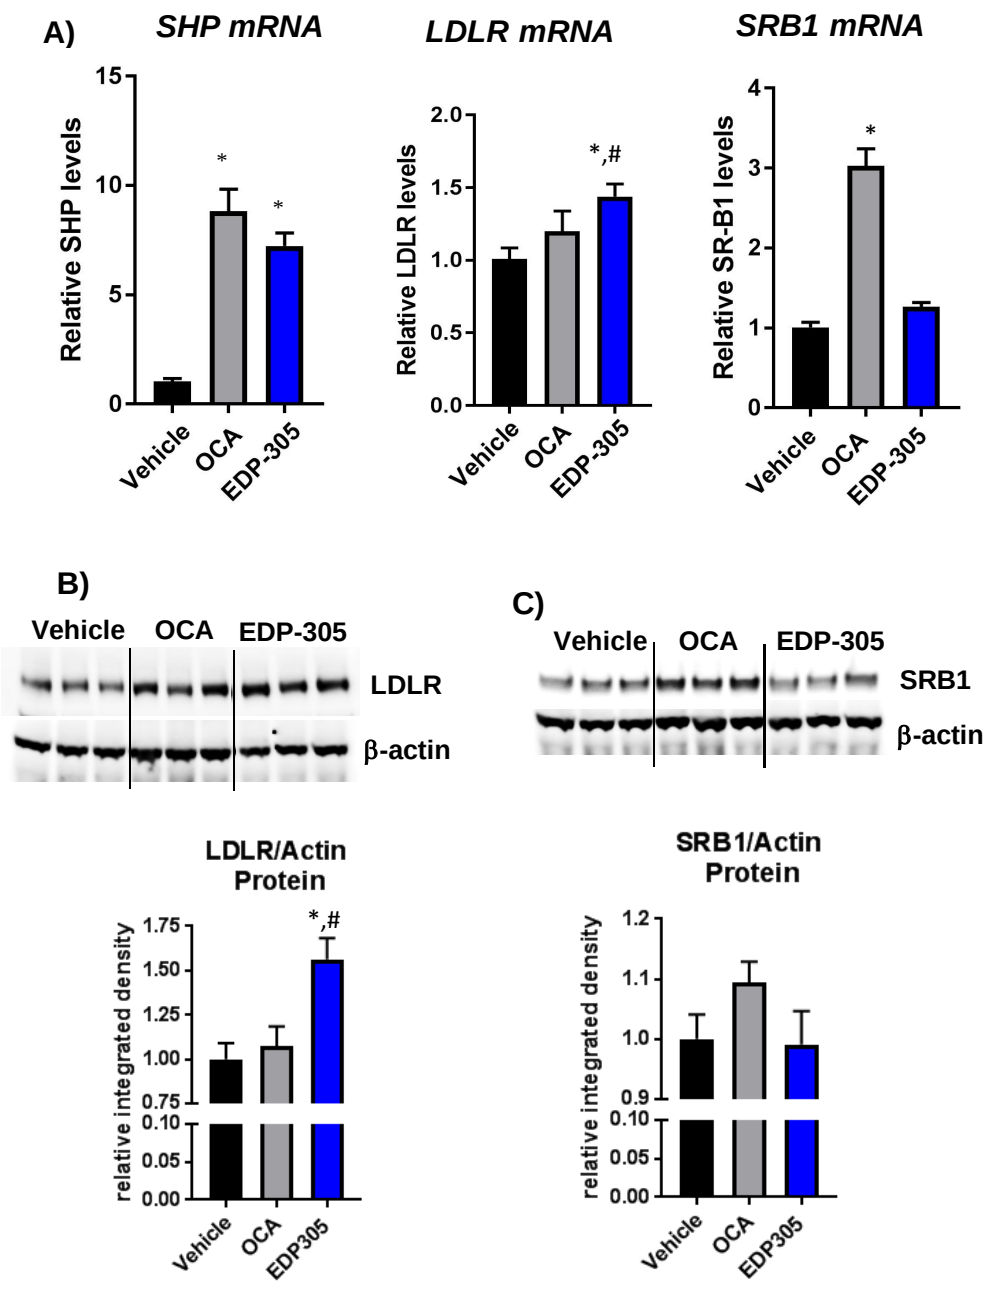
A)
SHP mRNA
LDLR mRNA
SRB1 mRNA
Relative SHP levels
Relative LDLR levels
Relative SR-B1 levels
Vehicle
OCA
EDP-305
B)
LDLR
β-actin
C)
SRB1
β-actin
LDLR/Actin Protein
SRB1/Actin Protein
relative integrated density
EDP305

Figure 6 A) Relative SHP, LDLR and SRB1 mRNA expression in C57Bl6 mice treated with compounds. Western blot and quantification of **B)** LDLR and **C)** SRB1 protein in liver tissue from EDP305 and OCA treated C57Bl6 mice. *p<0.05 relative to DMSO; #p<0.05 relative to EDP-305.

Conclusions

EDP-305 has distinct transcriptional and post-transcriptional regulatory mechanisms for LDLR and SRB1 expression when compared to OCA. The EDP-305 mechanisms driving these lipoprotein-related gene regulatory differences may translate to a more positive lipoprotein profile in patients, making EDP-305 attractive for further investigation in NASH.

Acknowledgement

- We thank Ruichao Shen for synthesizing the compounds.

Contact Information

To request a pdf of this poster, email **ljiang@enanta.com**