ENANTA Pharmaceuticals

#FRI-363

A novel and highly potent FXR agonist EDP-305 decreases liver steatosis, ballooning, and NAS score in a diet-induced murine model of non-alcoholic steatohepatitis (NASH)

Li-Juan Jiang[1], Mary Dinh Le Chau[1], Yang Li[1], Manuel Roqueta-Rivera[1], Guoqiang Wang[1], François Briand[2], Thierry Sulpice[2], Yat Sun Or[1]

[1]Enanta Pharmaceuticals, Inc. Watertown, MA, USA; [2] Physiogenex, Labege, France

To request a pdf of this poster, email **ljiang@enanta.com**

Introduction

Non-alcoholic steatohepatitis (NASH), characterized by steatosis, lobular inflammation, perisinusoidal fibrosis and hepatocyte ballooning, develops in a significant proportion of patients with non-alcoholic fatty liver disease (NAFLD). NASH is a leading cause of cirrhosis and hepatocellular carcinomas. NAFLD currently affects 20-40% of the general population, with 10-20% of those patients developing NASH. NASH is therefore becoming a major health issue in close association with obesity and diabetes[1]. There are currently no approved pharmacological therapies for NASH, underscoring the urgent need for effective treatments. The Farnesoid X Receptor (FXR) has emerged as an attractive target for NASH treatment due to its role in regulating glucose and lipid metabolism, as well as inflammatory responses in the liver[2]. EDP-305, a selective and potent small molecule FXR agonist, is currently in clinical trials for the treatment of NASH and primary biliary cholangitis (PBC).

The diet-induced NASH (DIN) model from Physiogenex (Laberge, France) mimics the human situation in which diet plays a major role in the development of NAFLD, resembling the human NAFLD / NASH physiopathological context. DIN mice, fed a high fat, high cholesterol, and high fructose diet, develop liver steatosis in 6 weeks, and early NASH associated with insulin resistance and obesity in 17 weeks. Herein we report the therapeutic efficacy of EDP-305 in DIN mice with NASH.

Methods

NASH was induced in male C57Bl/6 mice with a high fat/high cholesterol diet plus 10% fructose in drinking water (DIN). All treatments were administered between 6 weeks (steatohepatitis with incipient ballooning) and 16 weeks (advanced steatohepatitis with advanced ballooning) on DIN (n=10/group). Two doses of EDP-305 (10 and 30 mg/kg) or vehicle were administered with diet. A parallel group received OCA (30mg/kg) as a comparator. At 3, 6 and 10 weeks of treatment (i.e 9, 12 and 16 weeks of diet), blood was collected to measure serum chemistries. Liver injury and progression of NASH, including liver steatosis and ballooning, were evaluated by serum chemistry and histology, after 10 weeks of treatment.

Results

Fig 1. EDP-305 reduces plasma and liver lipid content in the DIN mouse. (A) Plasma total cholesterol, (B) Hepatic total cholesterol, (C) Triglycerides, and (D) Fatty acid levels in DIN mice treated with vehicle, EDP-305 10 mg/kg, EDP-305 30 mg/kg, and OCA 30 mg/kg.

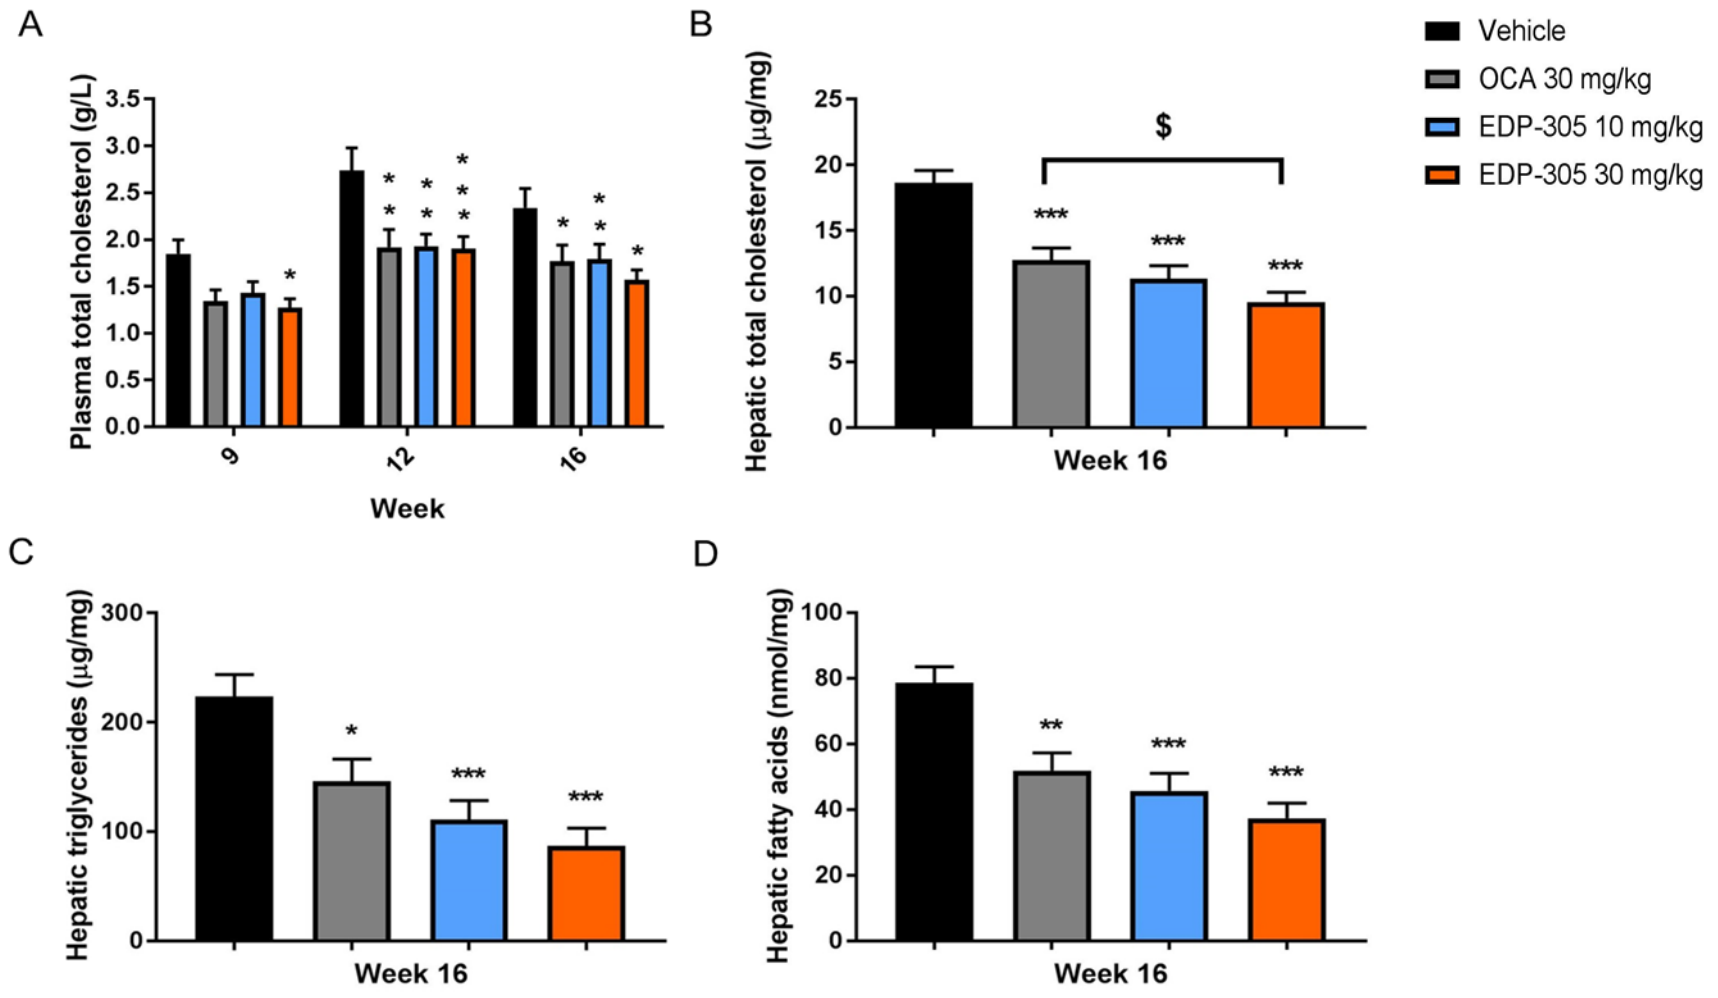
A
Plasma total cholesterol (g/L)
Week
9
12
16
B
Hepatic total cholesterol (µg/mg)
Week 16
C
Hepatic triglycerides (µg/mg)
Week 16
D
Hepatic fatty acids (nmol/mg)
Week 16
Vehicle
OCA 30 mg/kg
EDP-305 10 mg/kg
EDP-305 30 mg/kg

p<0.05, **p<0.01 and *p<0.001 vs. vehicle with two-way ANOVA + Bonferonni's;*

$ p<0.05 with one-way ANOVA + Newman-Keuls

Fig 2. EDP-305 decreases hepatocyte ballooning and NAFLD Activity Score (NAS). (A) Steatosis score; (B) Oil Red O staining; (C) Hepatocyte ballooning score; (D) NAS in DIN mice treated with vehicle, EDP-305 10 mg/kg, EDP-305 30 mg/kg, and OCA 30 mg/kg.

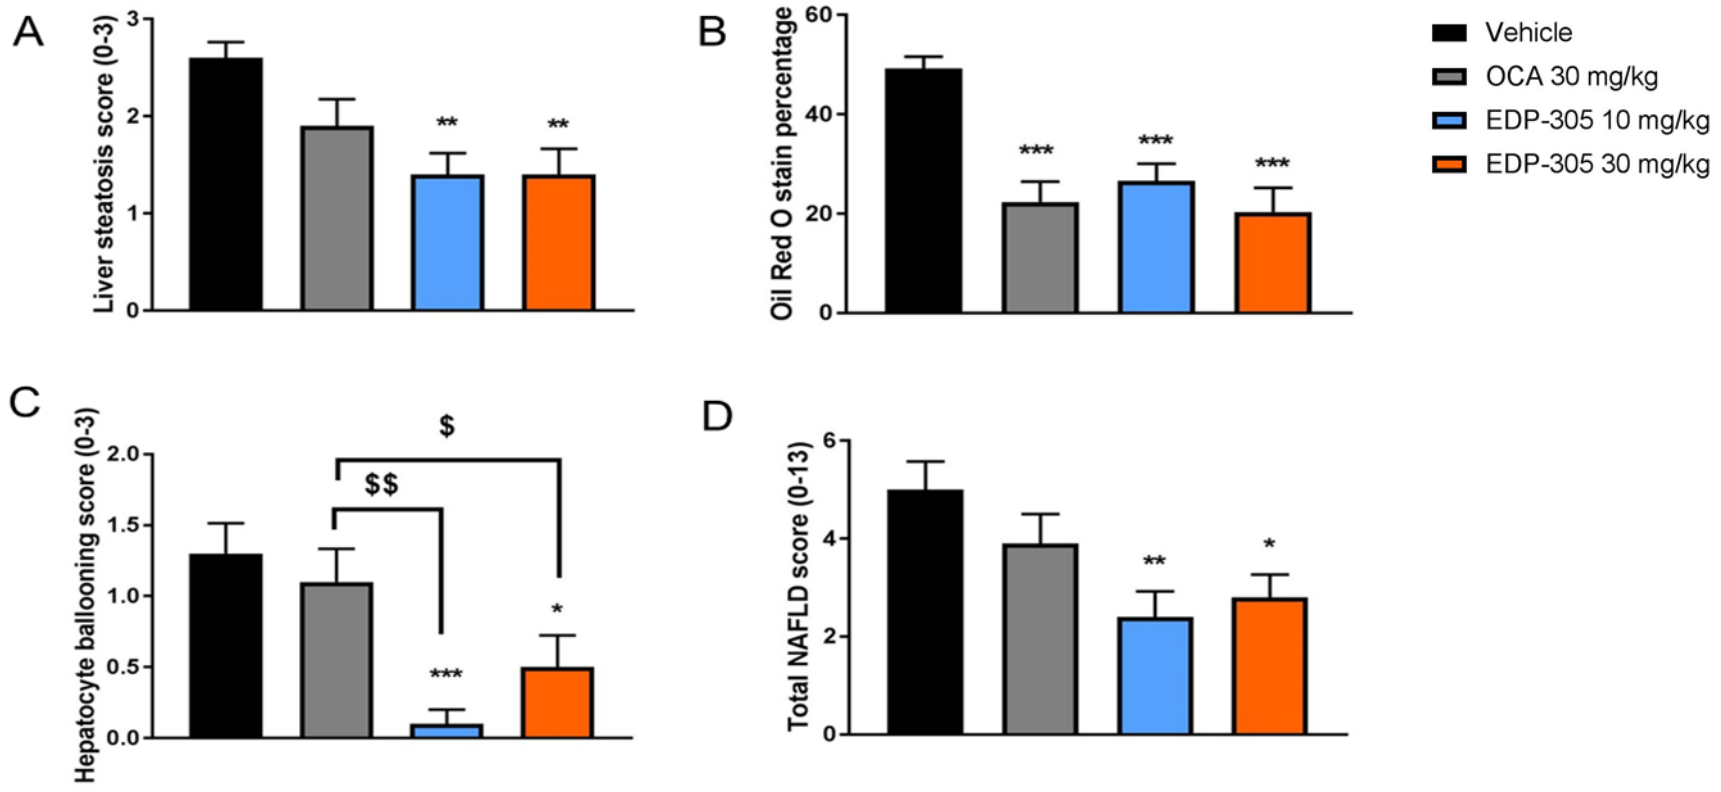
A
Liver steatosis score (0-3)
B
Oil Red O stain percentage
C
Hepatocyte ballooning score (0-3)
D
Total NAFLD score (0-13)
Vehicle
OCA 30 mg/kg
EDP-305 10 mg/kg
EDP-305 30 mg/kg

p<0.05, **p<0.01, *p<0.001 vs. vehicle with one way ANOVA + Dunnett's;*

$p<0.05 and $$p<0.001 with one way ANOVA + Newman-Keuls

Fig 3. EDP-305 decreases liver injury in DIN mice. Plasma ALT (A) and AST (B) levels in DIN mice treated with vehicle, EDP-305 10 mg/kg, EDP-305 30 mg/kg, and OCA 30 mg/kg.

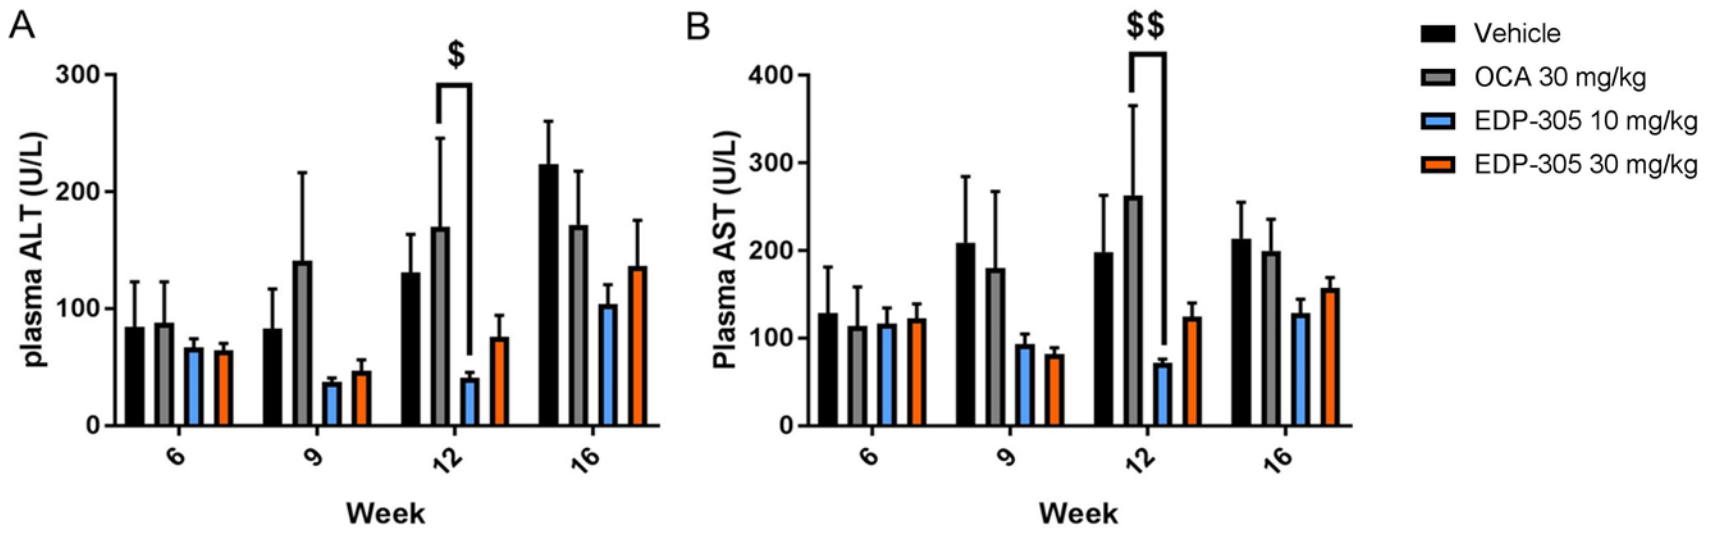
A
plasma ALT (U/L)
Week
6
9
12
16
B
Plasma AST (U/L)
Week
Vehicle
OCA 30 mg/kg
EDP-305 10 mg/kg
EDP-305 30 mg/kg

$ p<0.05 and $$ p<0.001 with Kruskal-Wallis + Dunnets post test

Fig 4. EDP-305 decreases inflammatory and fibrotic gene expression in DIN mice. Expression of TIMP-1 (A), α-SMA (B), MCP-1 (C), and TGF-β (D) in DIN mice treated with vehicle, EDP-305 at 10 or 30mg/kg, or OCA 30 mg/kg.

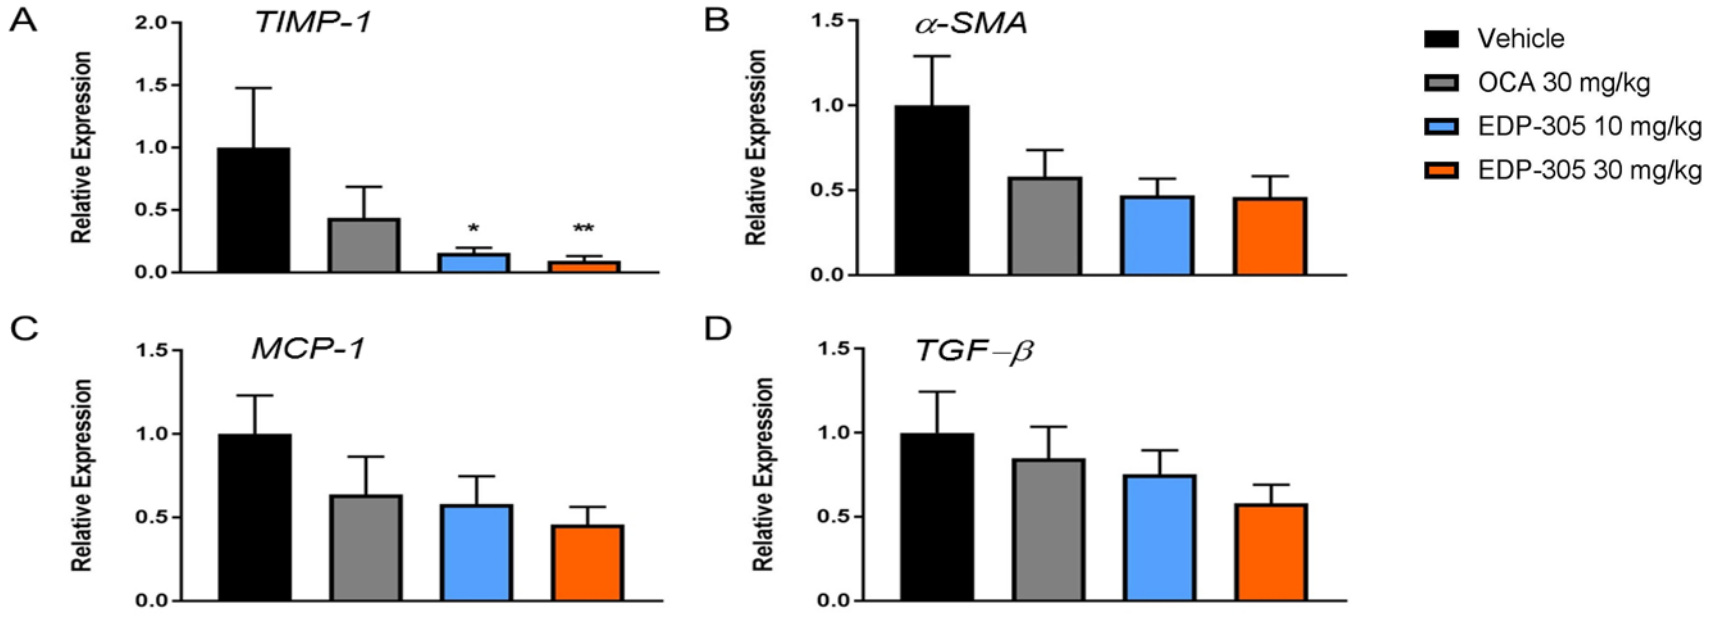
A
TIMP-1
Relative Expression
B
α-SMA
C
MCP-1
D
TGF–β
Vehicle
OCA 30 mg/kg
EDP-305 10 mg/kg
EDP-305 30 mg/kg

** p<0.05 and ** p<0.001 with Kruskal-Wallis + Dunnets post test*

Conclusions

EDP-305 exerts beneficial pharmacological effects in a dietary-induced NASH mouse model that mimics the human NAFLD/NASH physiological context. EDP-305 significantly decreased liver steatosis, hepatocyte ballooning, and total NAS score in DIN mice, suggesting that EDP-305 may have potential beneficial effects in treating NASH.

Acknowledgement

We thank Ruichao Shen for preparing all the compounds used in these studies.

References

1. Adorini, L et al. 2012. Farnesoid X receptor targeting to treat nonalcoholic steatohepatitis. *Drug Discov Today*. **17**: 988-97.
2. Schaap, F et al. 2014. Bile acid receptors as targets for drug development. *Nat Rev Gastroenterol Hepatol*. **11**: 55-67.

EASL, The International Liver Congress, 19-23 April, 2017, Amsterdam, The Netherlands

Enanta Pharmaceuticals, Inc.