ENANTA Pharmaceuticals

#1540

EDP-305, A Novel and Selective Farnesoid X Receptor Agonist, Exhibits Excellent Potency and Efficacy *In Vitro and In Vivo*

Yang Li, Mary Dinh Le Chau, Guoqiang Wang, Yat Sun Or, and Lijuan Jiang

Enanta Pharmaceuticals, Inc. Watertown, MA, USA

To request a pdf of this poster, email **ljiang@enanta.com**

Background

Nonalcoholic steatohepatitis (NASH) is becoming a major global health burden, with increasing prevalence and incidence worldwide. There is no approved therapy for the treatment of NASH. The farnesoid X receptor (FXR) has emerged as an important therapeutic target for the treatment of NASH [1]. EDP-305, a novel FXR agonist, was designed to meet this unmet medical need.

Methods

EDP-305 was tested side-by-side with obeticholic acid (OCA) for potency (EC_{50}) and efficacy using both a chimeric FXR reporter assay in Chinese hamster ovary (CHO) cells and a full length FXR luciferase reporter assay in human embryonic kidney (HEK) cells [2,3]. Activation of TGR5 was assessed by measuring the cellular level of cyclic adenosine monophosphate (cAMP) using a competitive immunoassay that quantitates based on enzyme fragment complementation. The ability of EDP-305 and OCA to regulate FXR downstream gene expression of small heterodimer partner (SHP), cholesterol 7 alpha-hydroxylase (CYP7A1) and bile salt export pump (BSEP) was tested in the human Huh7.5 hepatocyte cell line. The *in vivo* activity of EDP-305 and OCA was determined in C57BL/6 mice treated by oral gavage with EDP-305 or OCA at the indicated doses for five days.

Results

EDP-305 is a potent FXR agonist with an EC_{50} value of 8 nM in a full-length FXR reporter assay using HEK cells, which is a16-fold greater potency than OCA (EC_{50}: 130 nM). EDP-305 has minimal activity against TGR5 while OCA is active against TGR5. In addition, EDP-305 does not activate other nuclear receptors and is selective for FXR. Both EDP-305 and OCA showed dose-dependent increases in SHP and BSEP gene expression *in vitro*. At 12 nM, which is close to the EC_{50} of EDP-305 in the HEK cell reporter assay, EDP-305 induced SHP and BSEP mRNA expression by 5-fold and 18-fold, respectively, compared to minimal induction by OCA. EDP-305 reduced CYP7A1 mRNA expression down to approximately 5%, while 40% CYP7A1 mRNA remained with OCA treatment at 12 nM.

Consistent with the *in vitro* activation of FXR signaling, EDP-305 had similar effects *in vivo,* such that it induced a dose-dependent increase in the expression of target genes: SHP and fibroblast growth factor 15 (FGF15), in the mouse ileum. Moreover, EDP-305 treatment exhibited a dose-dependent increase in SHP and BSEP mRNA, and a dose-dependent reduction in CYP7A1 mRNA in the mouse liver.

Table 1. EDP-305 is Highly Selective for FXR

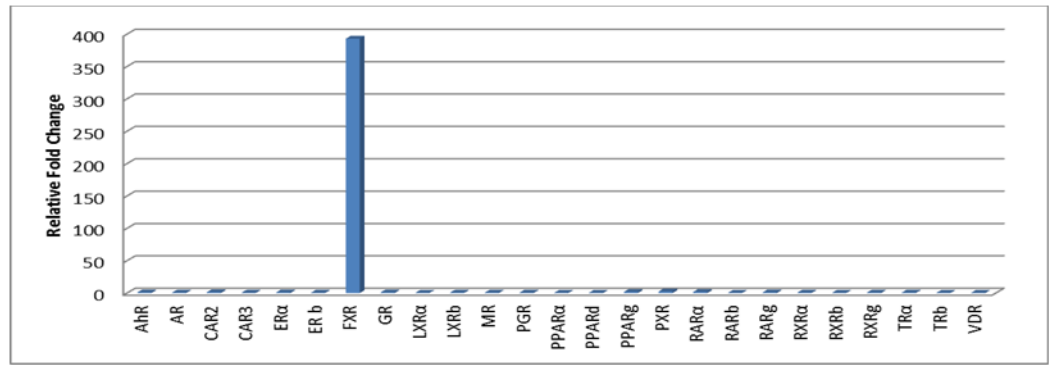

Relative Fold Change
400
350
300
250
200
150
100
50
0
AhR AR CAR2 CAR3 ERα ER b FXR GR LXRα LXRb MR PGR PPARα PPARd PPARg PXR RARα RARb RARg RXRα RXRb RXRg TRα TRb VDR

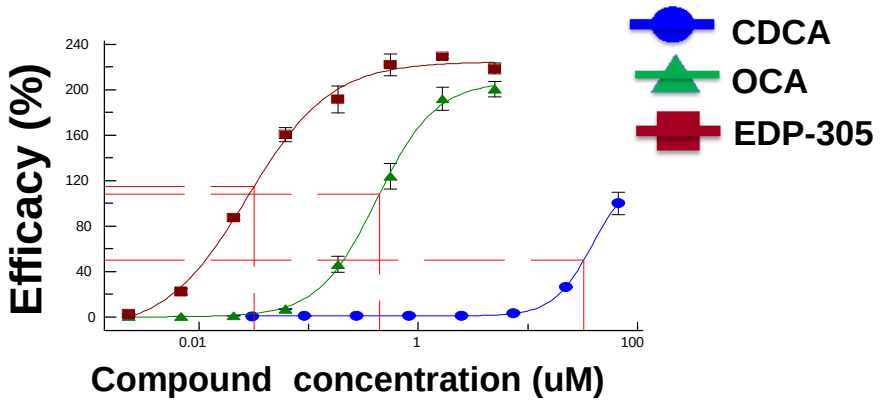

CDCA
OCA
EDP-305
Efficacy (%)
Compound concentration (uM)

Figure 1. Activation of FXR by EDP-305. Chimeric FXR and luciferase reporter constructs were transfected into CHO cells. Luciferase signal was measured after 22 hours of treatment with CDCA (chenodeoxycholic acid), OCA, or EDP-305.

Table 2. Potency and efficacy of EDP-305 and its metabolites in a chimeric FXR activation assay in CHO cells

Compound	Chimeric FXR activation (CHO)		
	N	EC50 (μM)	Efficacy (%)
CDCA	198	27.972 ± 0.915	100% ± 9%
OCA	198	0.569 ± 0.096	213% ± 39%
EDP-305	70	0.034 ± 0.008	223% ± 42%
2571 (EDP-305 Metabolite 1)	6	0.024 ± 0.011	195% ± 37%
2572 (EDP-305 Metabolite 2)	4	0.134 ± 0.037	176% ± 28%

Table 3. Potency and efficacy of EDP-305 and its metabolites in a full length FXR activation assay in HEK cells

Compound	Full length FXR activation (HEK)		
	N	EC50 (μM)	Efficacy (%)
CDCA	10	8.358 ± 1.392	100% ± 2%
OCA	10	0.130 ± 0.039	150% ± 21%
EDP-305	10	0.008 ± 0.003	152% ± 19%
2571 (EDP-305 Metabolite 1)	4	0.011 ± 0.012	149% ± 14%
2572 (EDP-305 Metabolite 2)	4	0.016 ± 0.019	151% ± 19%

Table 4. Potency and efficacy of EDP-305 and its metabolites in a TGR5 activation assay in CHO cells

Compound	TGR5 activation (CHO)		
	N	EC50 (μM)	Efficacy (%)
LCA (lithocholic acid)	56	0.129 ± 0.043	100% ± 9%
OCA	56	0.381 ± 0.102	72% ± 11%
EDP-305	14	>15	NA
2571 (EDP-305 Metabolite 1)	6	>15	NA
2572 (EDP-305 Metabolite 2)	4	>15	NA

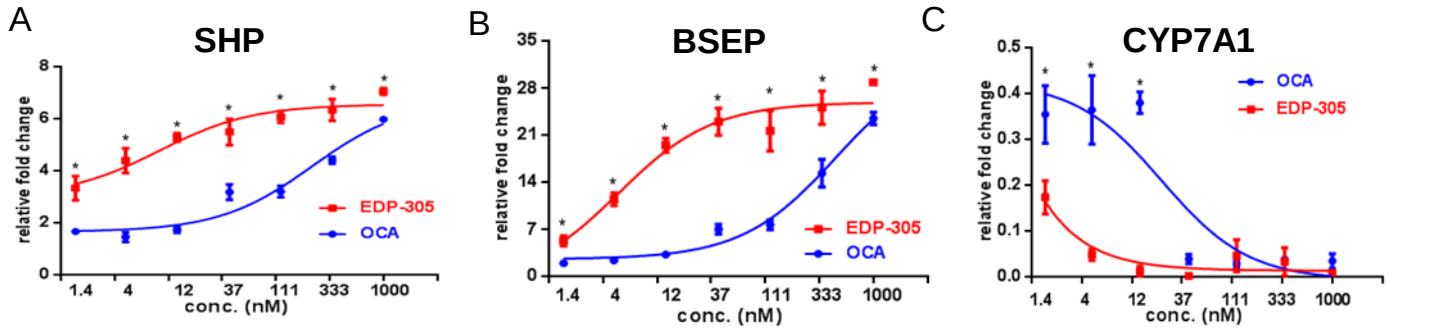

A
SHP
B
BSEP
C
CYP7A1
relative fold change
EDP-305
OCA
conc. (nM)

Figure 2. EDP-305 increases SHP, BSEP and CYP7A1 gene expression *in vitro*. HuH7.5 cells were seeded in tissue culture plates overnight in serum-reduced media (1% FBS). Cells were treated with EDP-305 or OCA for 10 hours (n=3). DMSO (0.2% v:v) served as a control. The highest treatment concentration for both EDP-305 and OCA were 1 uM with a series of 1:3 dilutions. (A) SHP; (B) BSEP; (C) CYP7A1. * *p<0.05 between EDP-305 and OCA.*

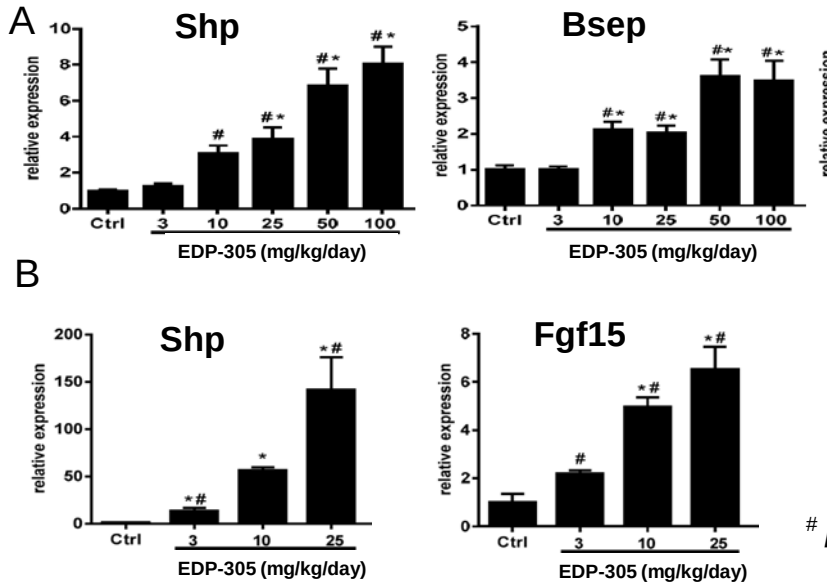

A
Shp
Bsep
Cyp7a1
B
Shp
Fgf15
relative expression
EDP-305 (mg/kg/day)

p<0.05 between EDP-305 and control.

Figure 3. EDP-305 regulates gene expression *in vivo.* C57/Bl6J mice, aged 8-10 weeks old, were treated with EDP-305. EDP-305 was administered via oral gavage. Gene expression analysis in liver (A) and intestine (B) were determined by real-time PCR. 18S ribosomal RNA (18S rRNA) was used as a house-keeping gene control.

Conclusions

EDP-305 is a highly potent and specific FXR agonist with minimal activity against TGR5. In contrast, OCA showed dual-agonist activity against both FXR and TGR5. EDP-305 demonstrated greater efficacy than OCA in the regulation of FXR target genes in bile acid metabolism both *in vitro* and *in vivo*.

Acknowledgements

We thank Ruichao Shen for preparing all the compounds used in these studies. We also thank Jun Zhang and Kristen Sagliani for their advice on the poster.

References

1. Adorini, L et al. 2012. Farnesoid X receptor targeting to treat nonalcoholic steatohepatitis. *Drug Discov Today*. **17**: 988-97
2. Molecular Pharmacology, 78:617–630, 2010
3. Chimeric FXR assay: http://indigobiosciences.com/products/fxr-nr1h4/

The 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11-15, 2016, Boston, MA, USA

Enanta Pharmaceuticals, Inc.